Number 507; Programme Code PEP6.8

Risk of Chronic Kidney Disease in Adult Patients With Chronic Hypoparathyroidism Treated With rhPTH(1-84) Compared With a Historical Control Cohort

Elvira O. Gosmanova,[1] Olulade Ayodele,[2] Nicole Sherry,[2] Fan Mu,[3] Allison Briggs,[3] Elyse Swallow,[3] Lars Rejnmark[4]

[1]Albany Medical College, Albany, NY, USA; [2]Shire Human Genetic Therapies, Inc., a Takeda company, Lexington, MA, USA; [3]Analysis Group, Inc., Boston, MA, USA; [4]Aarhus University and Aarhus University Hospital, Aarhus, Denmark

2 ways to access environmentally friendly e-posters

Scan the QR code

Access online: www.shirecongressposters.com/738691

No personal information is stored – standard charges may apply

BACKGROUND

- Patients with chronic hypoparathyroidism (HypoPT) are at increased risk of renal complications, including nephrocalcinosis, nephrolithiasis, and renal insufficiency[1-3]
- In long-term clinical trials, recombinant human parathyroid hormone (1-84), rhPTH(1-84), improved biochemical parameters and normalised urinary calcium excretion for patients with chronic HypoPT[4,5]
- In a previous study, estimated glomerular filtration rate (eGFR) was preserved among patients with chronic HypoPT treated with rhPTH(1-84) compared with patients not treated with rhPTH(1-84);[6] however, further evidence is required to establish whether treatment with rhPTH(1-84) reduces risk of renal complications in these patients

OBJECTIVE

- Evaluate chronic kidney disease (CKD) outcomes over a period of up to 5 years in adult patients with chronic HypoPT treated with rhPTH(1-84) in clinical trials compared with a historical control cohort of patients who did not receive rhPTH(1-84)

METHODS

- rhPTH(1-84)-treated patient cohort: derived from the REPLACE (NCT00732615), RELAY (NCT01268098), RACE (NCT01297309), and HEXT (NCT01199614) clinical trials (**Figure 1**)
- Historical control cohort: adult patients with chronic HypoPT selected from the large, nationally representative US Explorys electronic medical record database (Jan 2007–Aug 2019) using criteria similar to the trial enrolment criteria (**Figure 1**)
- Patients with CKD at baseline were excluded
- Analyses: Kaplan-Meier analysis and Cox proportional hazards models (see table and figure footnotes for additional analytic details)

Scan QR code for poster with additional details. Open downloaded poster and then click here

RESULTS

- Study cohorts: rhPTH(1-84), n=118; historical control cohort, n=478 (**Figure 2**)
- Kaplan-Meier analysis: rhPTH(1-84)-treated patients had a significantly reduced risk of developing CKD during follow-up compared with patients in the control cohort not treated with rhPTH(1-84) (*P*<0.01; **Figure 3**)
- Multivariable adjusted Cox proportional hazards model: hazard ratio for developing CKD associated with rhPTH(1-84) compared with no rhPTH(1-84) treatment was 0.47 (95% CI: 0.25–0.88; *P*<0.05; **Table 1**)

Figure 1. Patient Selection Flow Diagram

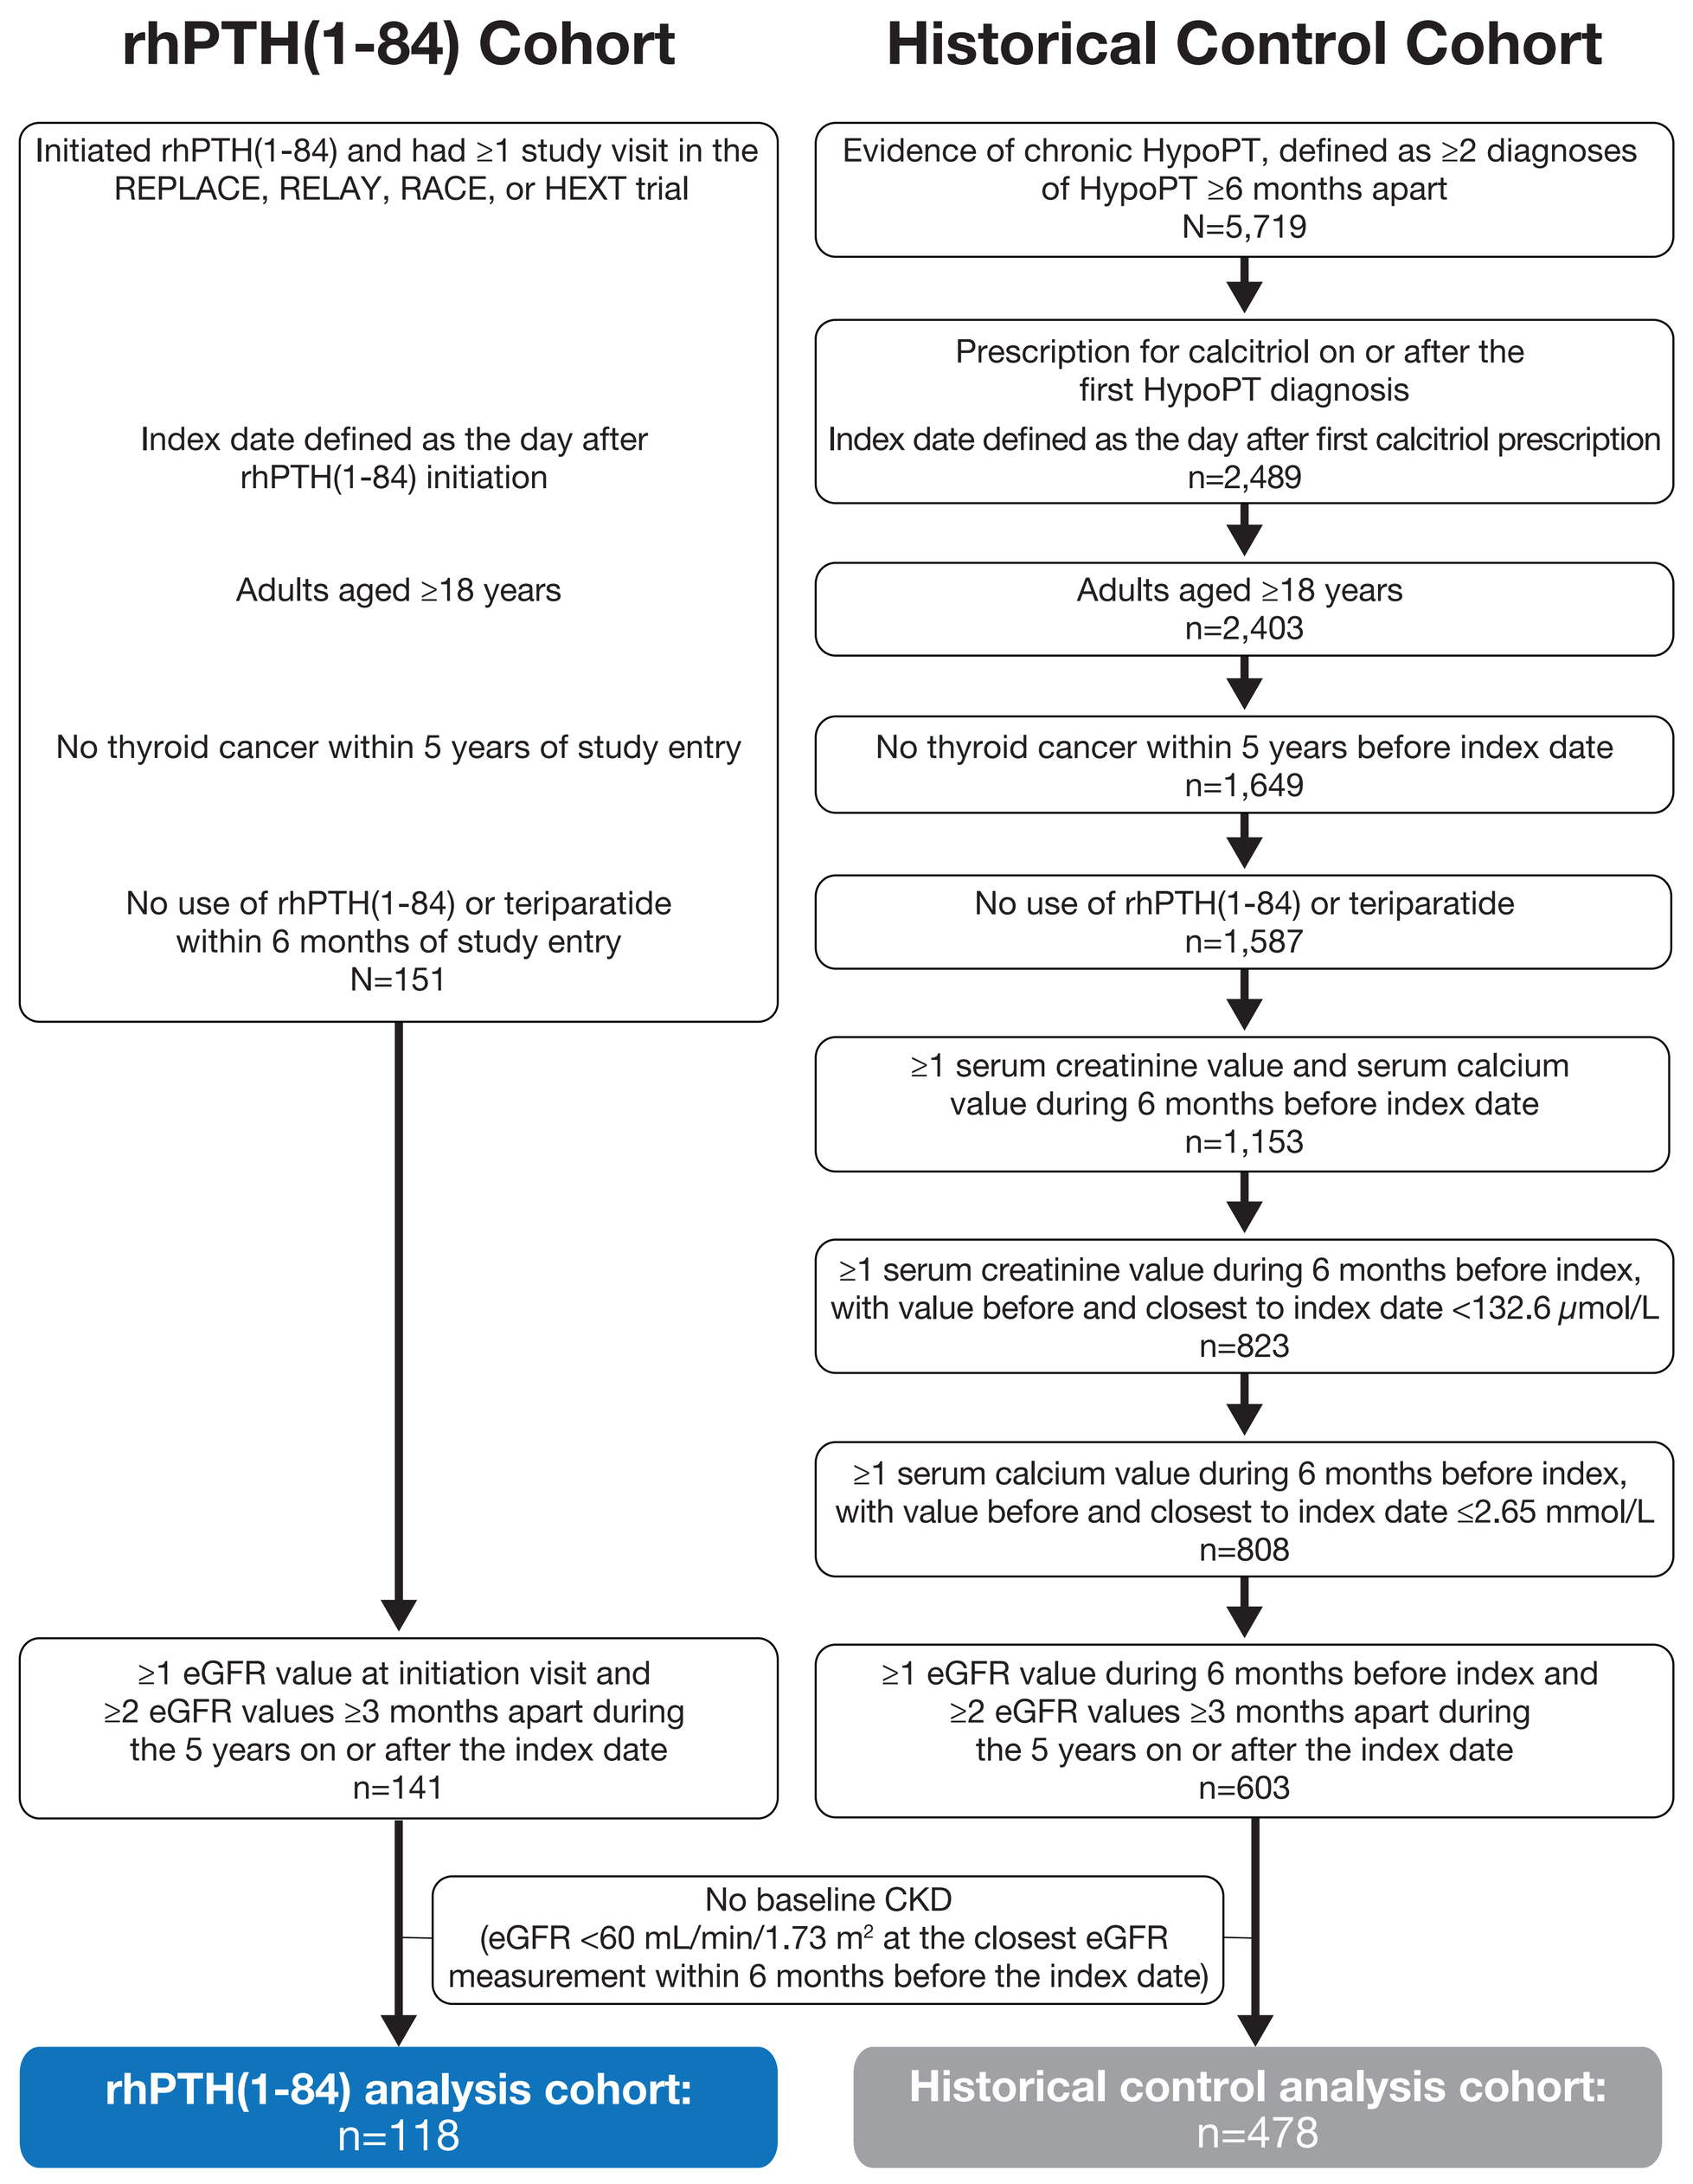

rhPTH(1-84) Cohort
Historical Control Cohort
Initiated rhPTH(1-84) and had ≥1 study visit in the REPLACE, RELAY, RACE, or HEXT trial
Index date defined as the day after rhPTH(1-84) initiation
Adults aged ≥18 years
No thyroid cancer within 5 years of study entry
No use of rhPTH(1-84) or teriparatide within 6 months of study entry N=151
Evidence of chronic HypoPT, defined as ≥2 diagnoses of HypoPT ≥6 months apart N=5,719
Prescription for calcitriol on or after the first HypoPT diagnosis Index date defined as the day after first calcitriol prescription n=2,489
Adults aged ≥18 years n=2,403
No thyroid cancer within 5 years before index date n=1,649
No use of rhPTH(1-84) or teriparatide n=1,587
≥1 serum creatinine value and serum calcium value during 6 months before index date n=1,153
≥1 serum creatinine value during 6 months before index, with value before and closest to index date <132.6 μmol/L n=823
≥1 serum calcium value during 6 months before index, with value before and closest to index date ≤2.65 mmol/L n=808
≥1 eGFR value at initiation visit and ≥2 eGFR values ≥3 months apart during the 5 years on or after the index date n=141
≥1 eGFR value during 6 months before index and ≥2 eGFR values ≥3 months apart during the 5 years on or after the index date n=603
No baseline CKD (eGFR <60 mL/min/1.73 m² at the closest eGFR measurement within 6 months before the index date)
rhPTH(1-84) analysis cohort: n=118
Historical control analysis cohort: n=478

eGFR=estimated glomerular filtration rate; HypoPT=hypoparathyroidism.

Figure 2. Baseline Patient Demographic and Clinical Characteristics

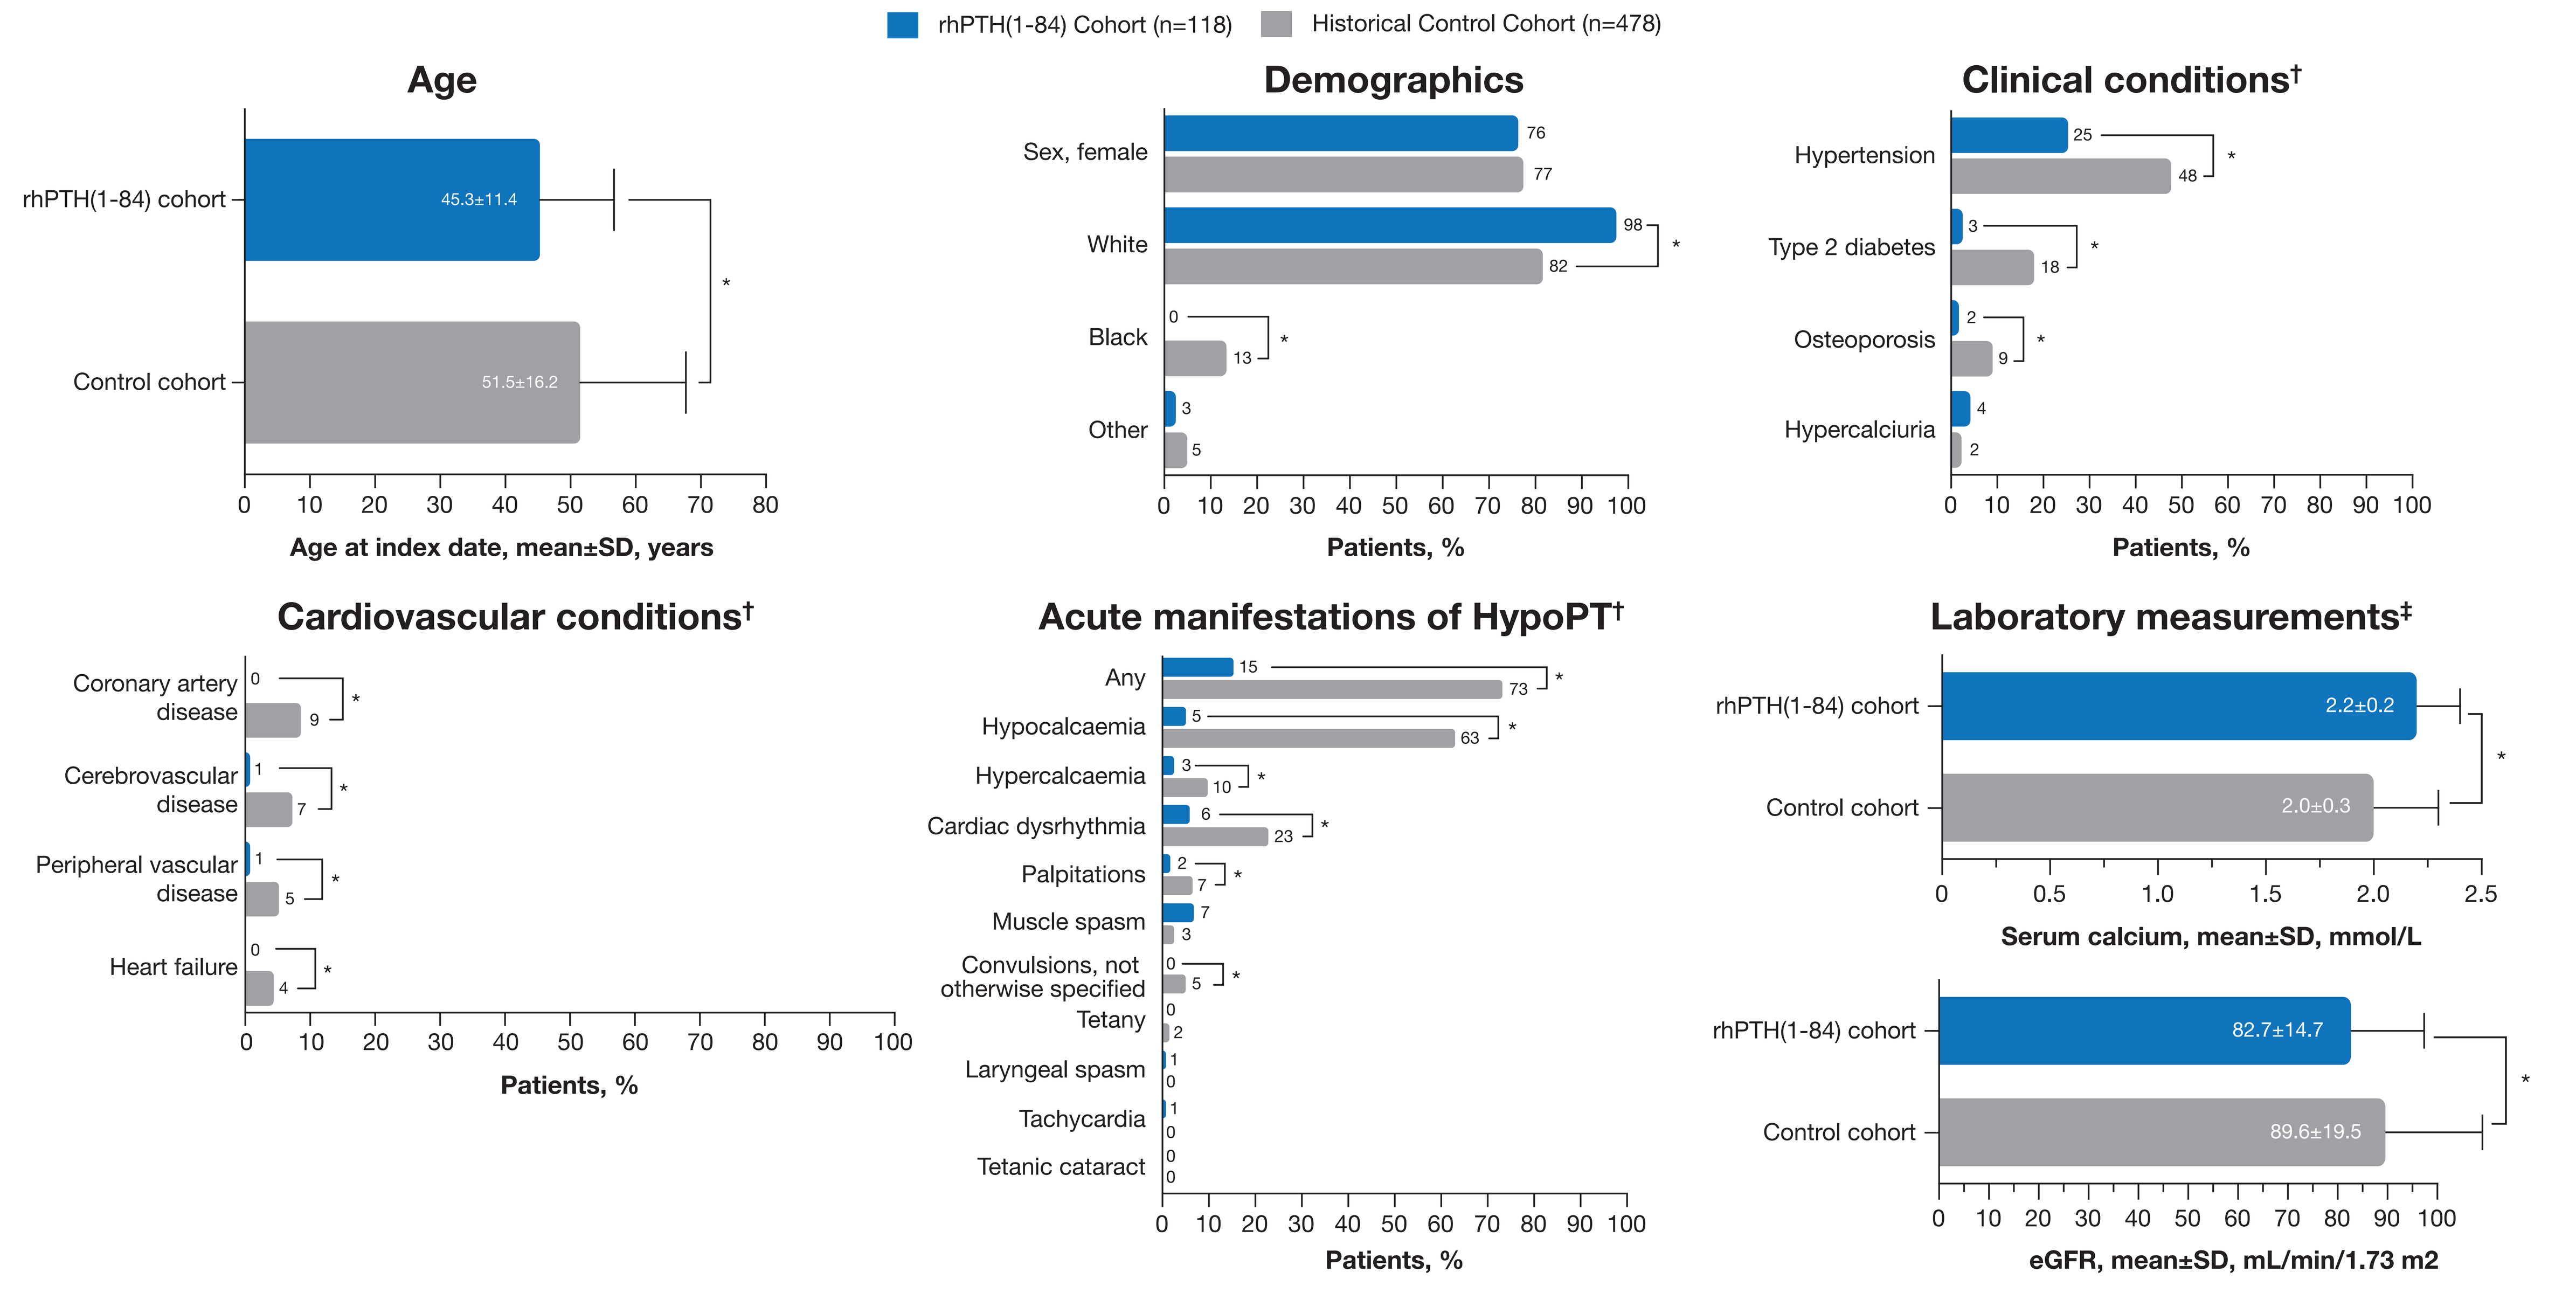

rhPTH(1-84) Cohort (n=118)
Historical Control Cohort (n=478)
Age
rhPTH(1-84) cohort 45.3±11.4
Control cohort 51.5±16.2
Age at index date, mean±SD, years
Demographics
Sex, female 76 77
White 98 82
Black 0 13
Other 3 5
Patients, %
Clinical conditions†
Hypertension 25 48
Type 2 diabetes 3 18
Osteoporosis 2 9
Hypercalciuria 4 2
Patients, %
Cardiovascular conditions†
Coronary artery disease 0 9
Cerebrovascular disease 1 7
Peripheral vascular disease 1 5
Heart failure 0 4
Patients, %
Acute manifestations of HypoPT†
Any 15 73
Hypocalcaemia 5 63
Hypercalcaemia 3 10
Cardiac dysrhythmia 6 23
Palpitations 2 7
Muscle spasm 7 3
Convulsions, not otherwise specified 0 5
Tetany 0 2
Laryngeal spasm 1 0
Tachycardia 1 0
Tetanic cataract 0 0
Patients, %
Laboratory measurements‡
rhPTH(1-84) cohort 2.2±0.2
Control cohort 2.0±0.3
Serum calcium, mean±SD, mmol/L
rhPTH(1-84) cohort 82.7±14.7
Control cohort 89.6±19.5
eGFR, mean±SD, mL/min/1.73 m2

eGFR=estimated glomerular filtration rate; HypoPT=hypoparathyroidism.
**P*<0.05.
†Before index date; medical terms defined by diagnosis or procedure codes.
‡Closest measurement before index date.

CKD outcome: First occurrence of eGFR <60 mL/min/1.73 m² on or after the index date, confirmed by a second measurement ≥3 months later

Figure 3. Kaplan-Meier Analysis of Time to First Instance of CKD Among Patients With HypoPT Without CKD at Baseline

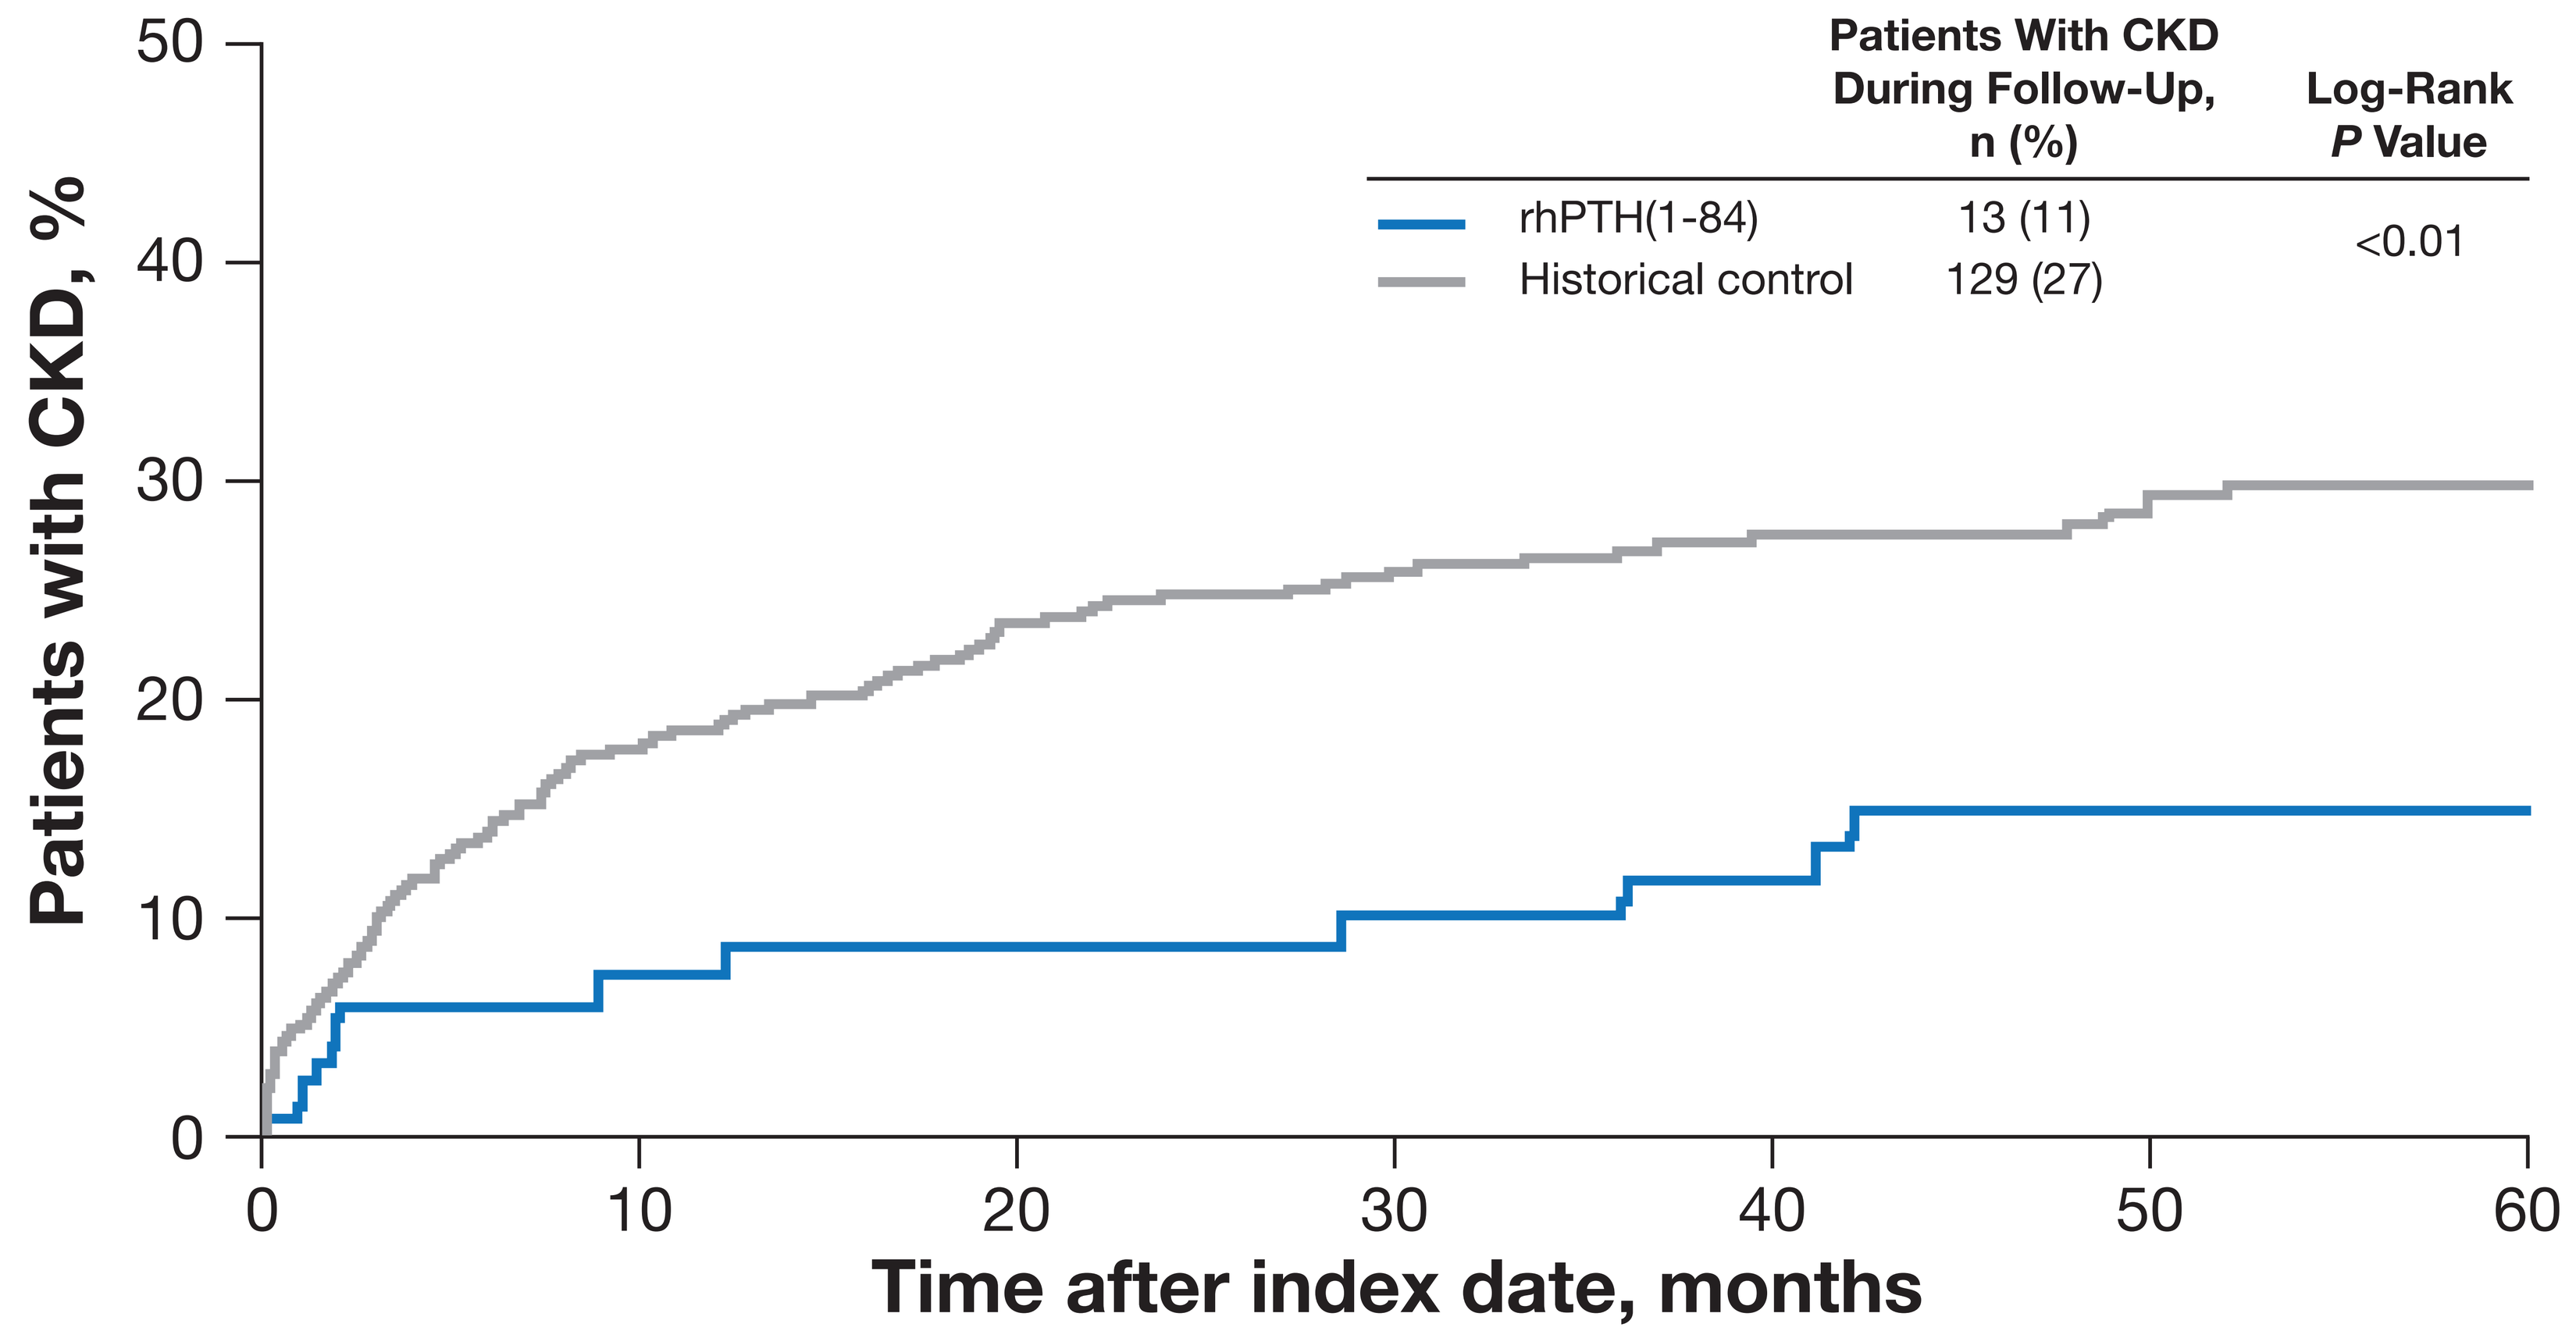

Patients With CKD During Follow-Up, n (%)
Log-Rank P Value
rhPTH(1-84) 13 (11)
Historical control 129 (27)
<0.01
Patients with CKD, %
Time after index date, months

	0	10	20	30	40	50	60
Number of patients with CKD							
rhPTH(1-84) patients	0	8	9	10	11	13	13
Historical control patients	0	85	110	119	124	128	129
Number of patients at risk							
rhPTH(1-84) patients	118	66	63	59	56	53	48
Historical control patients	478	381	307	259	202	151	121

CKD=chronic kidney disease; eGFR=estimated glomerular filtration rate; HypoPT=hypoparathyroidism.

Table 1. Risk of Developing CKD

Cox Proportional Hazards Model of Time to First Instance of CKD Among Patients With Chronic HypoPT and Without CKD at Baseline			
Estimated effect of rhPTH(1-84) vs historical control cohort			
	HR	**95% CI**	***P* Value**
Unadjusted	0.43	0.24–0.76	<0.01
Adjusted†	0.47	0.25–0.88	<0.05

CI=confidence interval; CKD=chronic kidney disease; CV=cardiovascular; eGFR=estimated glomerular filtration rate; HR=hazard ratio; HypoPT=hypoparathyroidism.
†Cox models were adjusted for demographics (age, sex, race), clinical conditions (hypercalciuria, hypertension, type 2 diabetes, CV conditions, and acute manifestations of HypoPT), and laboratory values (eGFR and serum calcium) at baseline.
CV conditions were defined as ≥1 diagnosis of cerebrovascular disease, coronary artery disease, heart failure, or peripheral vascular disease; medical terms defined by diagnosis or procedure codes.
Acute manifestations of HypoPT were defined as ≥1 diagnosis of hypercalcaemia, hypocalcaemia, laryngeal spasm, muscle spasm, convulsions (not otherwise specified), tetanic cataract, tetany, cardiac dysrhythmia, tachycardia, or palpitations; medical terms defined by diagnosis or procedure codes.

Patients with chronic HypoPT treated with **rhPTH(1-84)** had reduced risk of developing CKD over 5 years compared with **control** patients not treated with rhPTH(1-84)

LIMITATIONS

- Differences in patient management between the studied cohorts (ie, predefined clinical trial protocols for the rhPTH(1-84) cohort vs real-world practice for the control cohort)
- Potential for miscoding of diagnoses and misreporting of laboratory test measurements in the administrative claims database

CONCLUSIONS

- **After adjusting for baseline differences, patients with chronic HypoPT treated with rhPTH(1-84) in clinical trials had a significantly reduced risk of developing CKD compared with patients in a control cohort who did not receive rhPTH(1-84)**
- **Further research is warranted to better understand the mechanisms underlying these findings**

REFERENCES

Scan QR code for poster with additional details. Open downloaded poster and then click here

DISCLOSURES

EOG has served as a consultant for Shire, a Takeda company. OA and NS are employees of Shire Human Genetic Therapies, Inc., a Takeda company, Lexington, MA, USA. FM, AB, and ES are employees of Analysis Group, Inc., which was contracted by Shire Human Genetic Therapies, Inc., a Takeda company, to conduct this research. LR has served as an advisory board member, speaker, and research investigator for Shire, a Takeda company. This study was funded by Shire Human Genetic Therapies, Inc., a Takeda company, Lexington, MA, USA. Editorial support was provided by ICON, North Wales, PA, USA, and was funded by Shire Human Genetic Therapies, Inc., a Takeda company, Lexington, MA, USA.

e-poster presented virtually at the European Society of Endocrinology Congress; 22–26 May, 2021

This poster is intended for healthcare providers